IRX4204 enhances gait recovery in mice subjected to experimental autoimmune encephalomyelitis

Gracious D. S. Kasheke,[1] Scott P. Holman,[1] Kaitlyn Fraser,[1] Basmah Hendy,[1] Arul Jude Cline Asainayagam,[1] Sagar Vuligonda,[2] Martin Sanders,[2] Antoine Hakim,[3] Marianna Foldvari,[3] and George S. Robertson [1,4]

[1]Department of Pharmacology and Brain Repair Centre, Faculty of Medicine, Dalhousie University, Life Sciences Research Institute, Halifax, Nova Scotia, Canada
[2] Io Therapeutics Inc., Santa Ana, California, USA
[3]School of Pharmacy, Faculty of Science, University of Waterloo, Waterloo, Ontario, Canada
[4]Department of Psychiatry, 5909 Veterans' Memorial Lane, 8th Floor, Abbie J. Lane Memorial Building, QEII Health Sciences Centre, Halifax, Nova Scotia, Canada

Introduction

Current immune-based therapies for multiple sclerosis (MS) reduce relapses but have limited value in slowing disease progression. Remyelination is considered essential for functional recovery in MS.[1]

Activation of the retinoid X receptor (RXR) enhances remyelination by promoting the differentiation of oligodendrocyte progenitor cells into myelin-producing oligodendrocytes.[2] IRX4204 is an investigational drug that preferentially activates the RXR resulting in a complex transcriptional response which resolves inflammation and promotes neural repair. Oral administration of IRX4204 been shown to reduce motor impairments in patients suffering from Parkinson's disease.[3]

We show that the oral administration of IRX4204 beginning at peak disease (day post immunization 16; DPI 16) reduced clinical scores, gait deficits, white matter loss, and axonal injury and enhanced remyelination in female mice subjected to experimental autoimmune encephalomyelitis (EAE). To improve the safety and efficacy of IRX4204, we have developed an intranasal nanoparticle formulation of IRX4204 that enables this drug to be preferentially delivered to the central nervous system (CNS)..

IRX4204 Reduces EAE Disease Severity

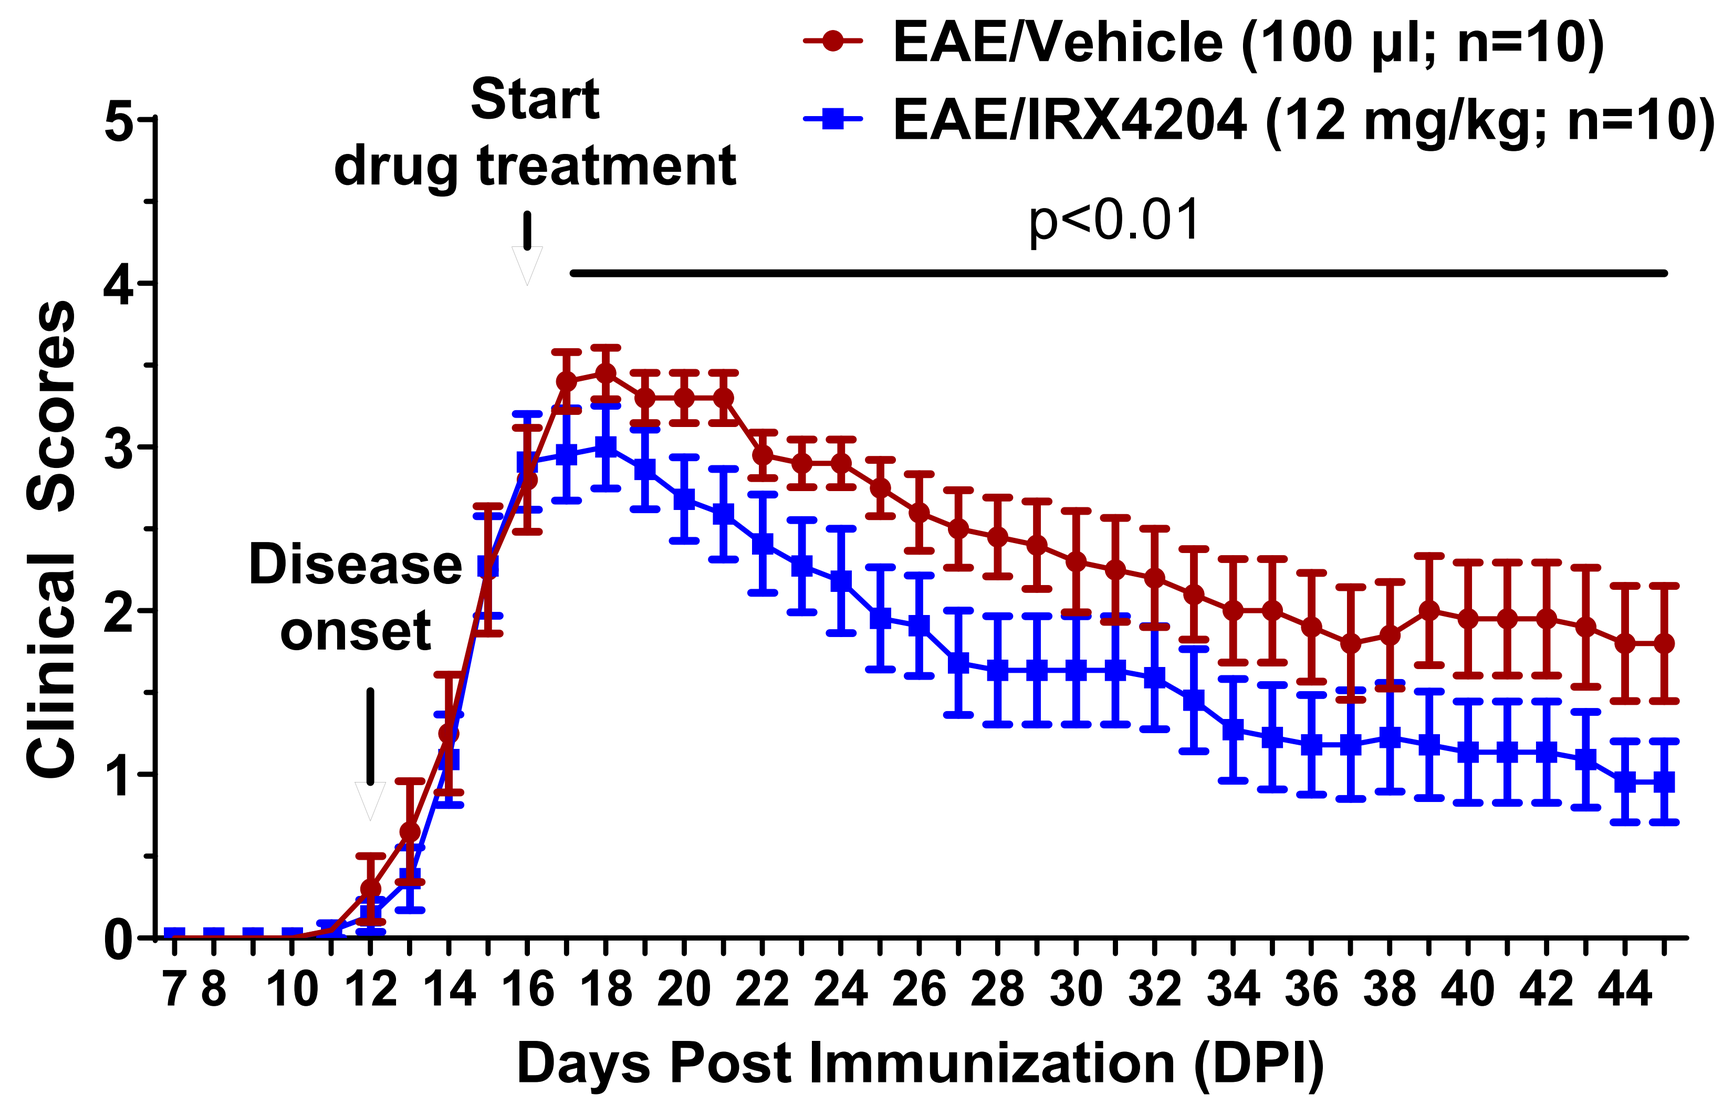
EAE/Vehicle (100 µl; n=10)
EAE/IRX4204 (12 mg/kg; n=10)
Start drug treatment
p<0.01
Disease onset
Clinical Scores
Days Post Immunization (DPI)

Clinical scores for EAE mice treated orally with vehicle or IRX4204 once daily beginning at DPI 16 and until DPI 44 (mean ± SEM). The clinical scores were lower for EAE/IRX4204 than EAE/Vehicle mice.

IRX4204 Reverses Gait Deficits in EAE Mice

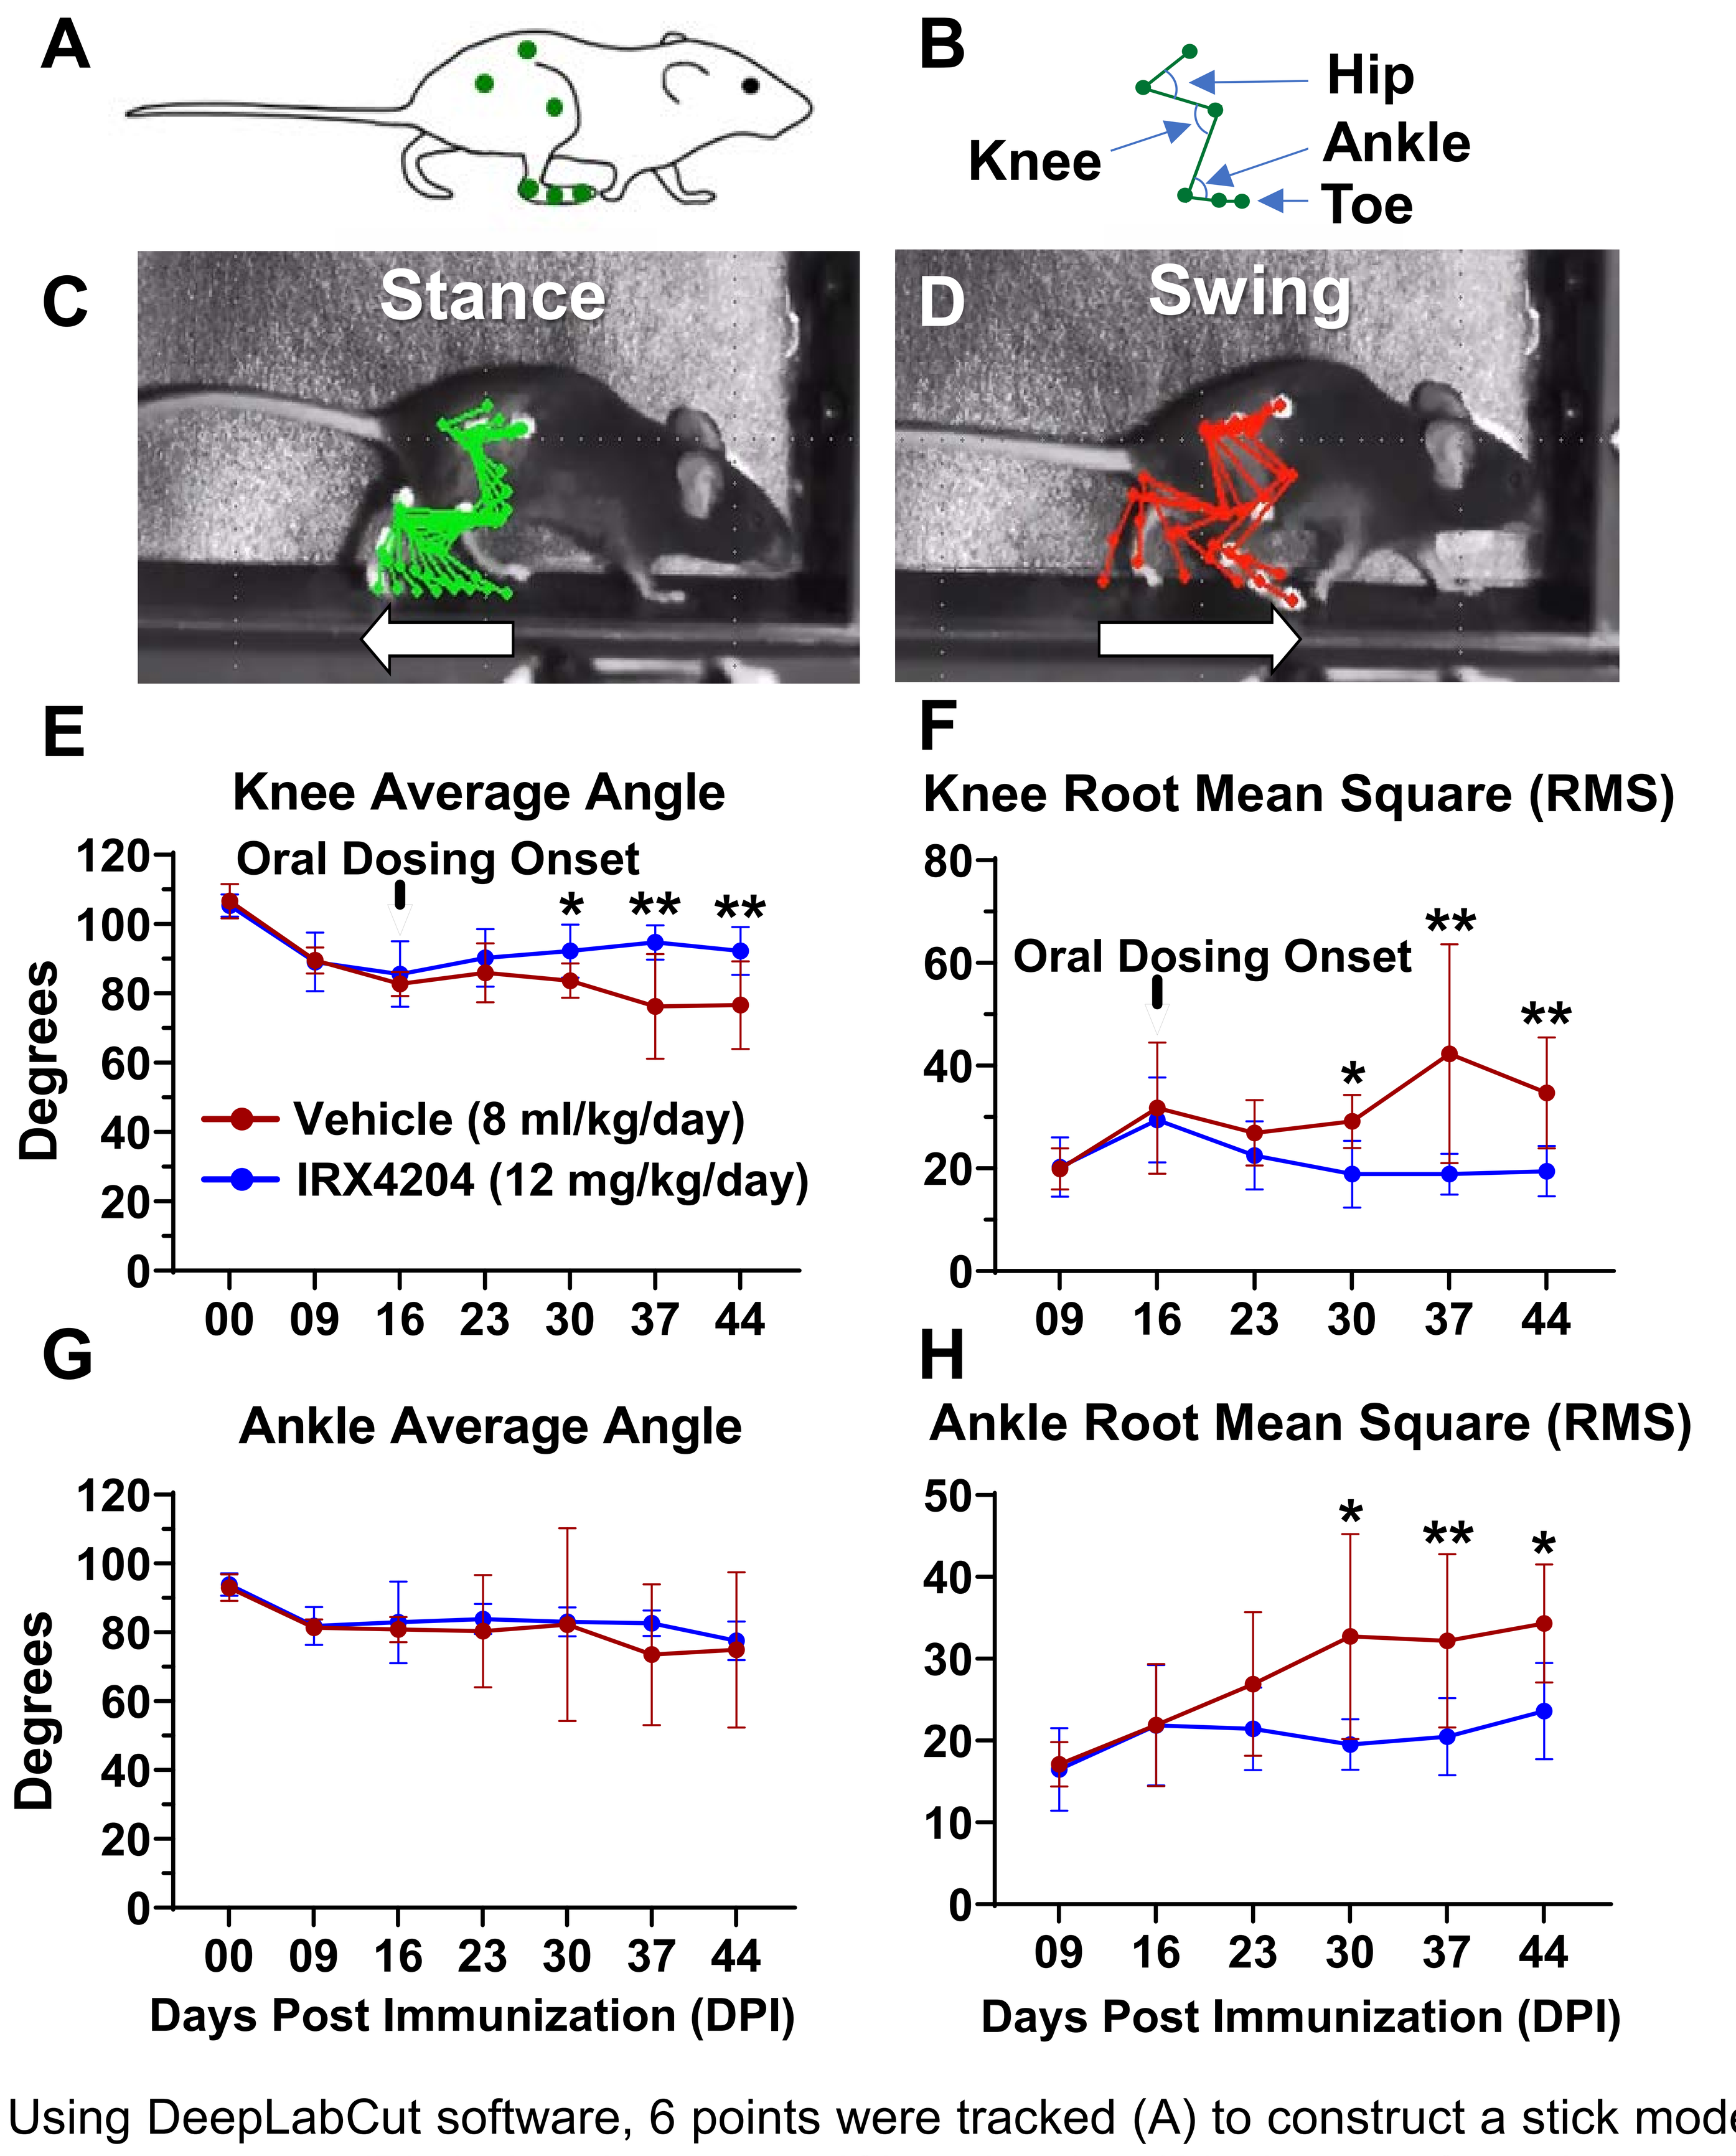
A
B
Hip
Knee
Ankle
Toe
C
Stance
D
Swing
E
Knee Average Angle
Oral Dosing Onset
Degrees
Vehicle (8 ml/kg/day)
IRX4204 (12 mg/kg/day)
F
Knee Root Mean Square (RMS)
Oral Dosing Onset
G
Ankle Average Angle
Degrees
H
Ankle Root Mean Square (RMS)
Days Post Immunization (DPI)
Days Post Immunization (DPI)

Using DeepLabCut software, 6 points were tracked (A) to construct a stick model for leg movements (B) during the stance (C) and swing (D) phases of a gait cycle while female mice walked on treadmill. Gait was measured 2 days before immunization with MOG35-55 (DPI 0) and DPI 9-44. Oral IRX4204 (12 mg/kg/day) dosing starting at peak disease (DPI 16) reversed the loss of knee average angle and elevation of RMS values and halted ankle RMS elevations from DPI 30-44 (E-H). Each point and bar represent the mean ± SD of data for 6 mice, *p<0.05, **p<0.01.

IRX4204 Enhances Remyelination and Reduces Axon Injury

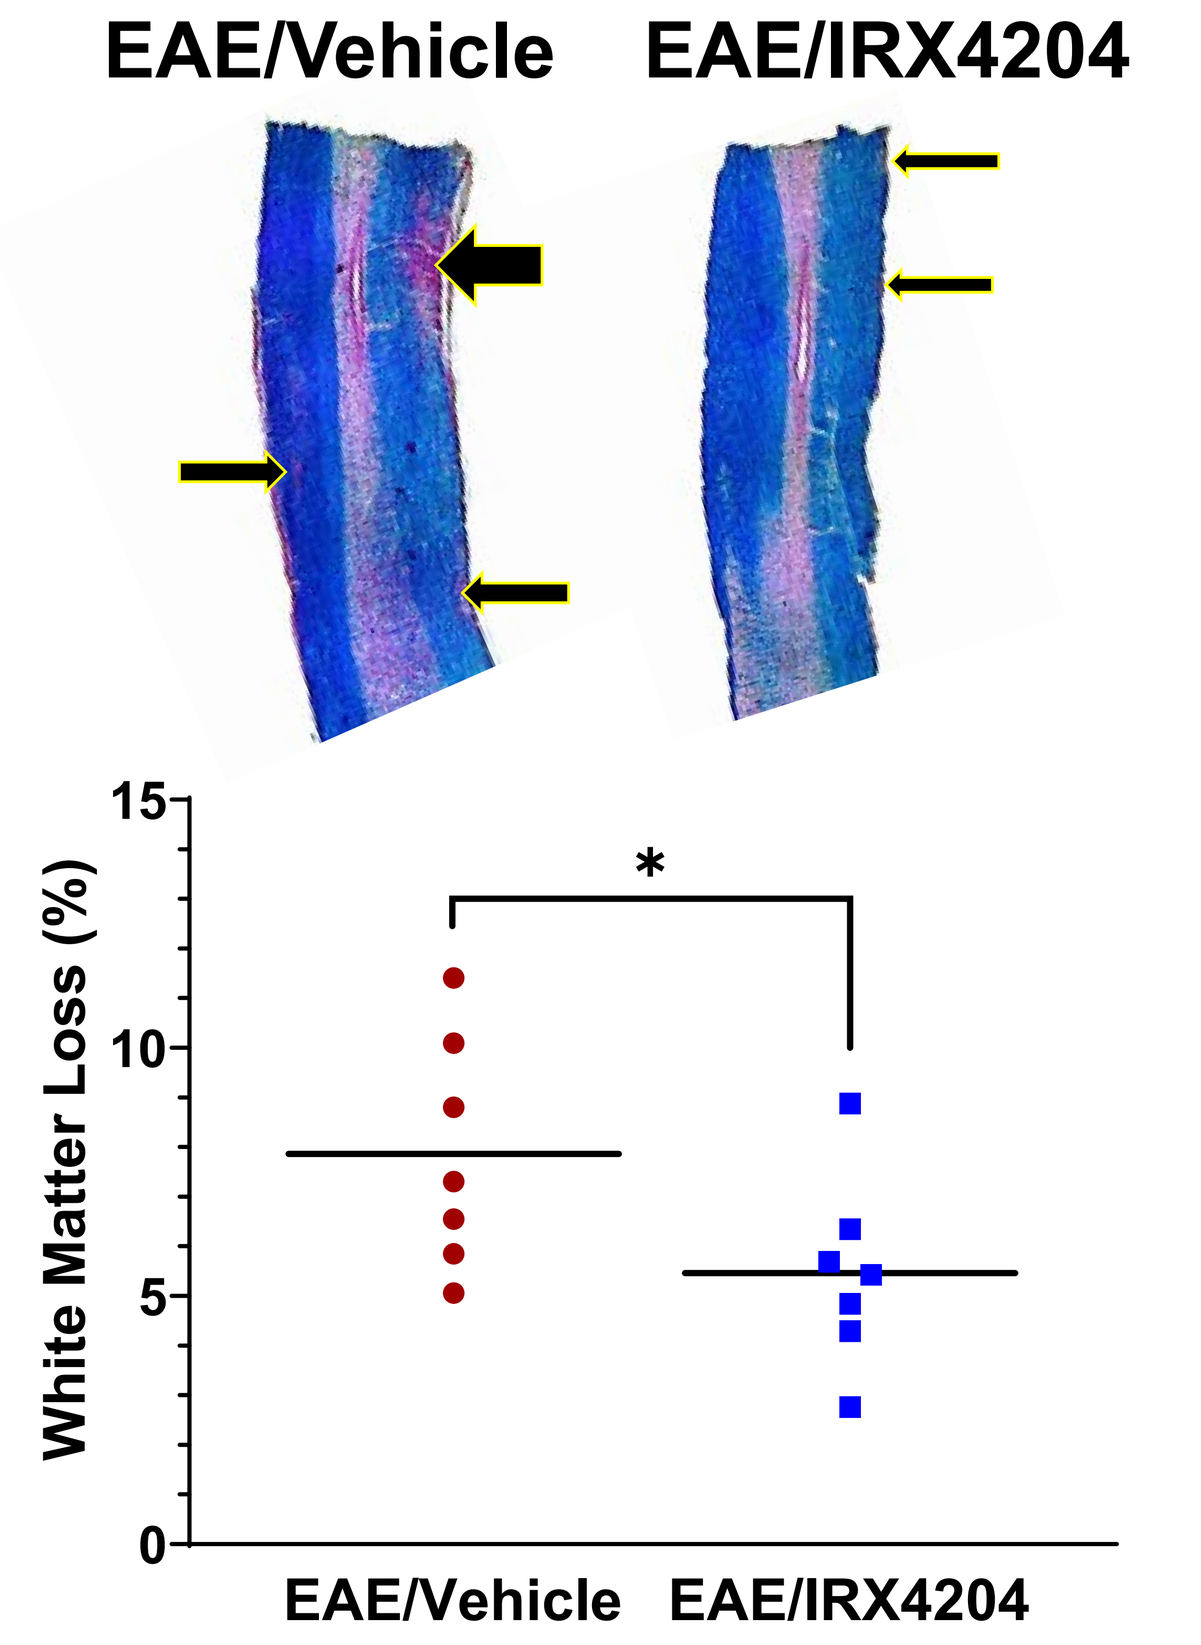
EAE/Vehicle
EAE/IRX4204
White Matter Loss (%)
EAE/Vehicle
EAE/IRX4204

Representative spinal cord sections from EAE mice treated orally with vehicle or IRX4204 stained with eriochrome cyanine. Quantification of eriochrome cyanine staining showed that IRX4204 reduced spinal cord white matter loss.

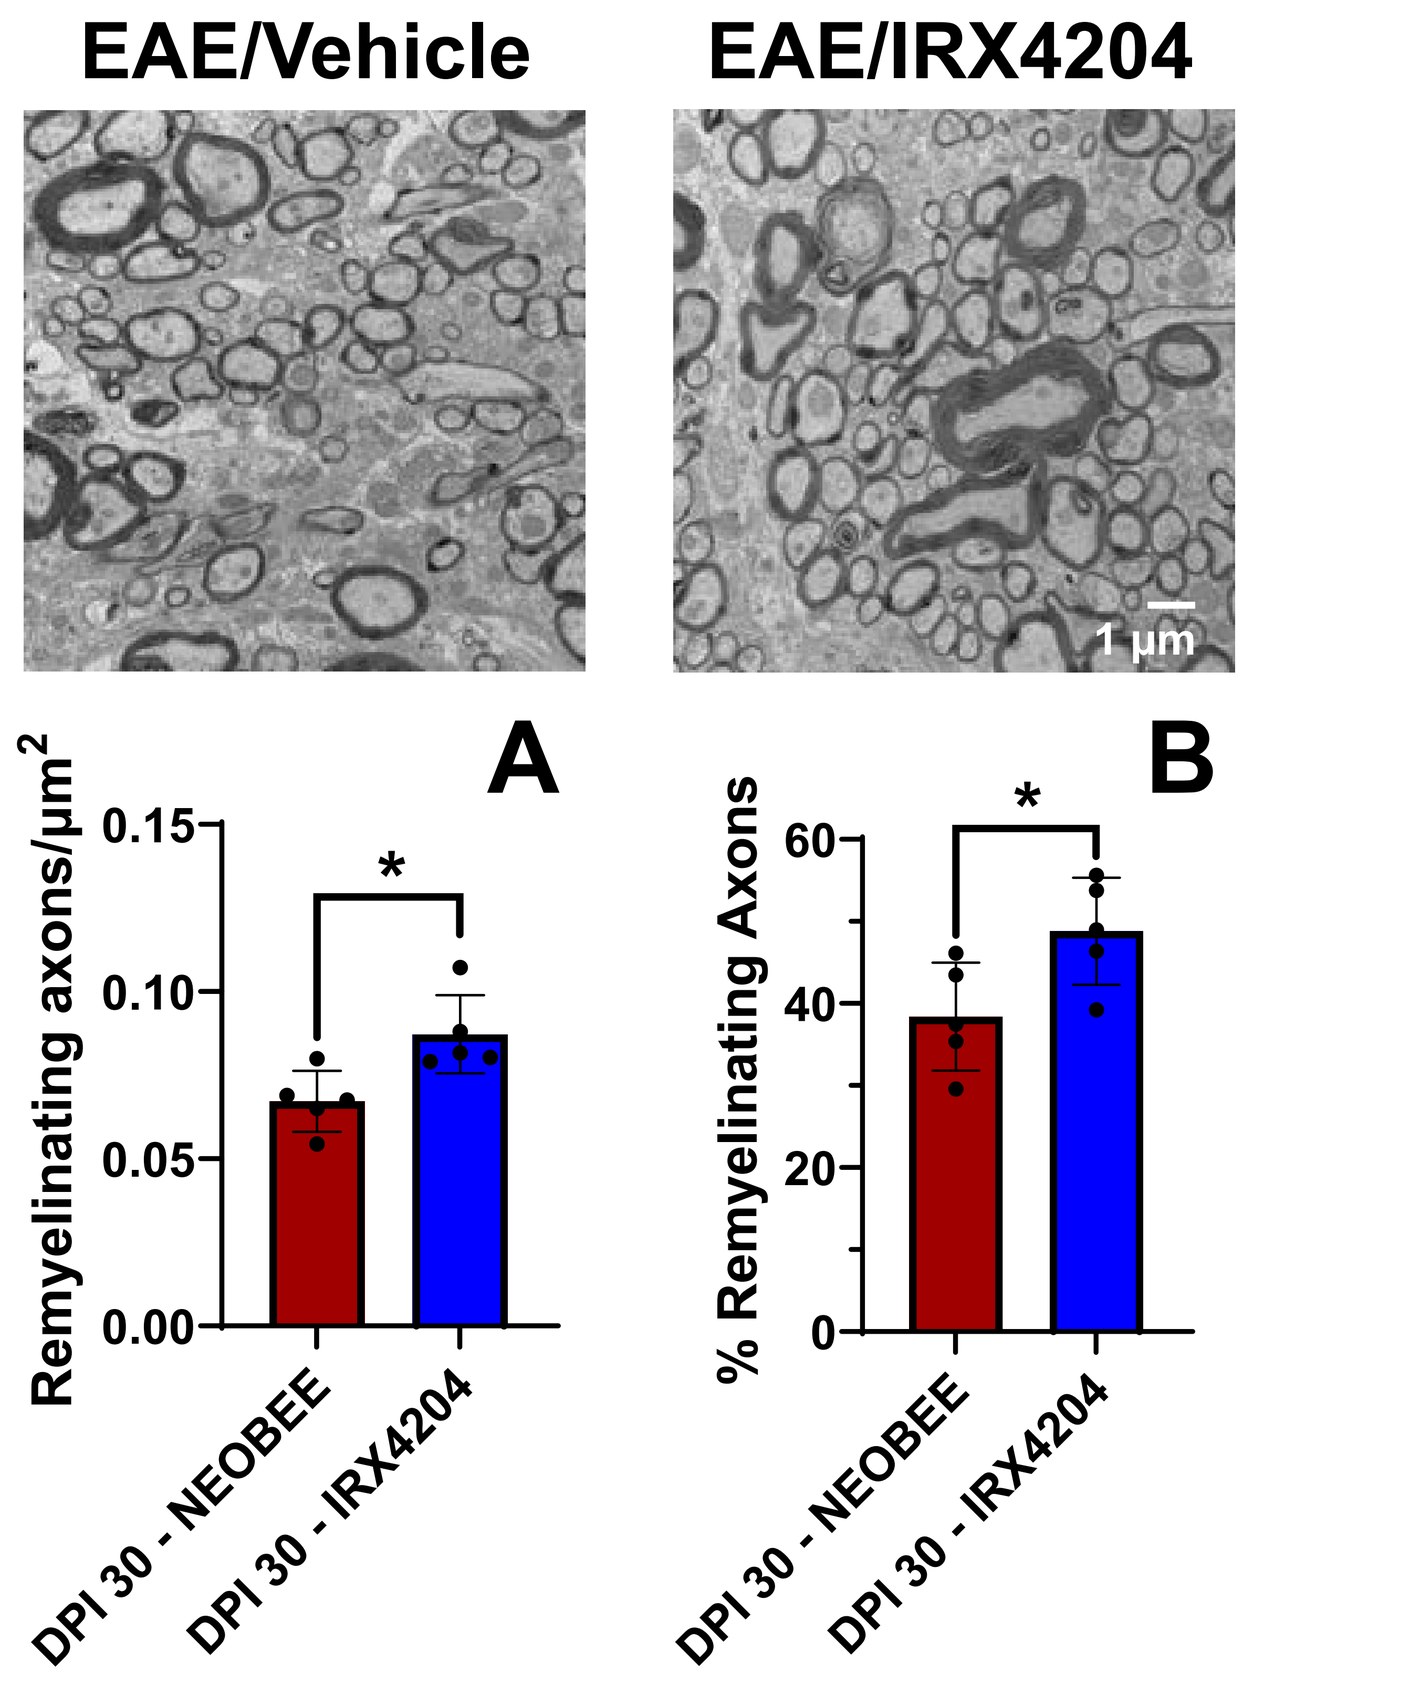
EAE/Vehicle
EAE/IRX4204
1 µm
A
Remyelinating axons/µm²
DPI 30 - NEOBEE
DPI 30 - IRX4204
B
% Remyelinating Axons
DPI 30 - NEOBEE
DPI 30 - IRX4204

Representative electron microscopy images of the L3/L4 spinal cord region of EAE mice treated orally with vehicle (NEOBEE) or IRX4204. Quantification of the number of remyelinating axons/µm² (A) and the percentage of remyelinating axons per image (14 x 14 µm in size; B) indicated that IRX4204 increased remyelination.

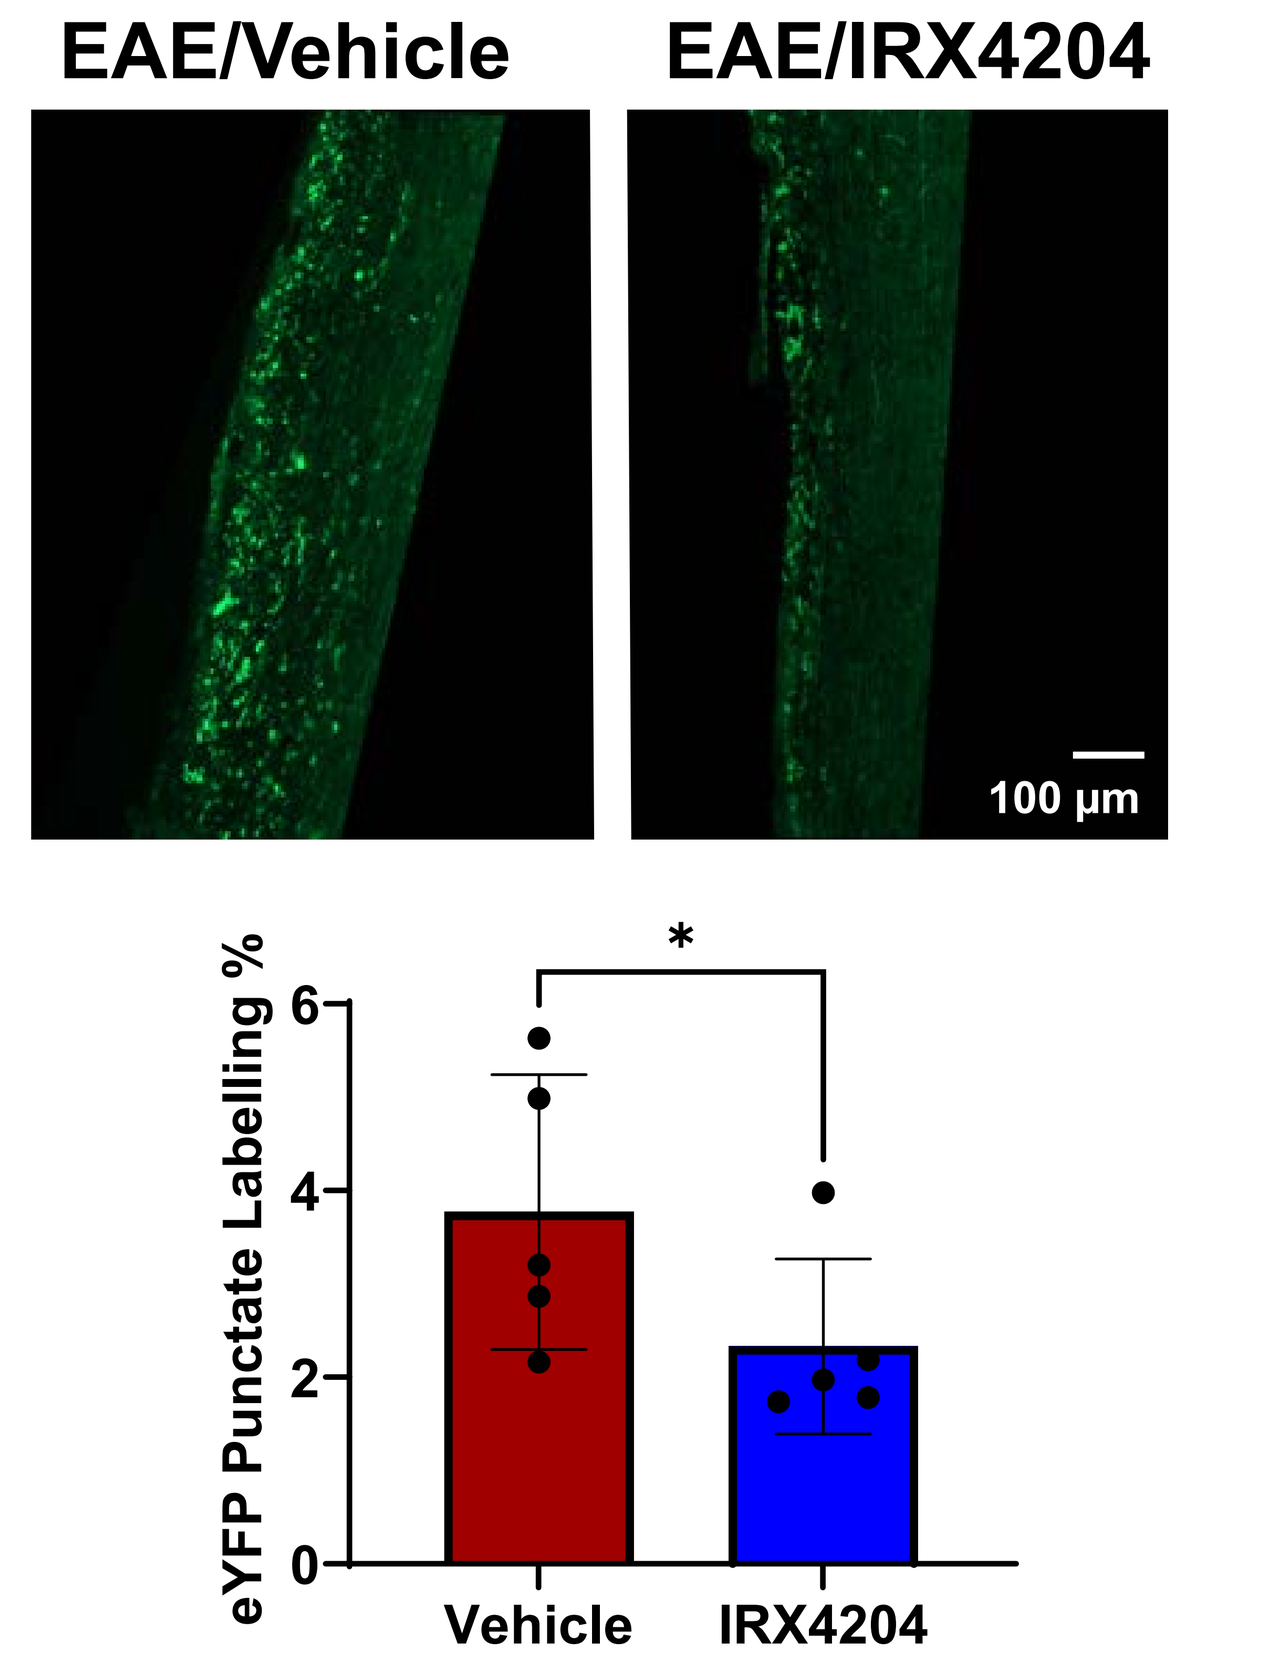
EAE/Vehicle
EAE/IRX4204
100 µm
eYFP Punctate Labelling %
Vehicle
IRX4204

Representative spinal cord (L1-L5) images of Thy1-eYFP reporter mice subjected to EAE and treated with either vehicle or IRX4204. Note that the reduction of punctate eYFP fluorescence in EAE mice that received IRX4204 indicative of decreased axonal transection.

Intranasal Delivery of IRX4204 Nanoparticles Produces a Sustained and Preferential Elevation of IRX4204 Concentrations and Pharmacodynamic Actions in the CNS

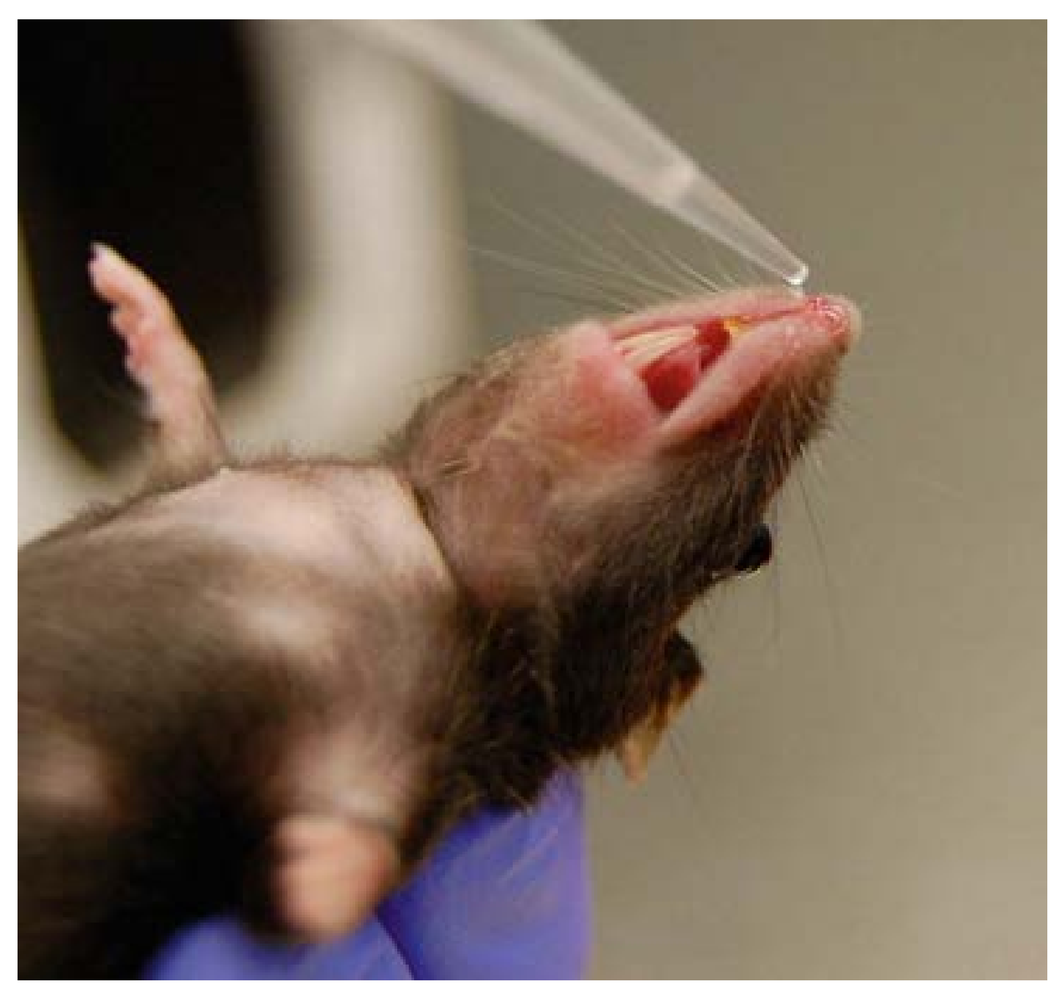

Intranasal Drug Delivery in the Mouse

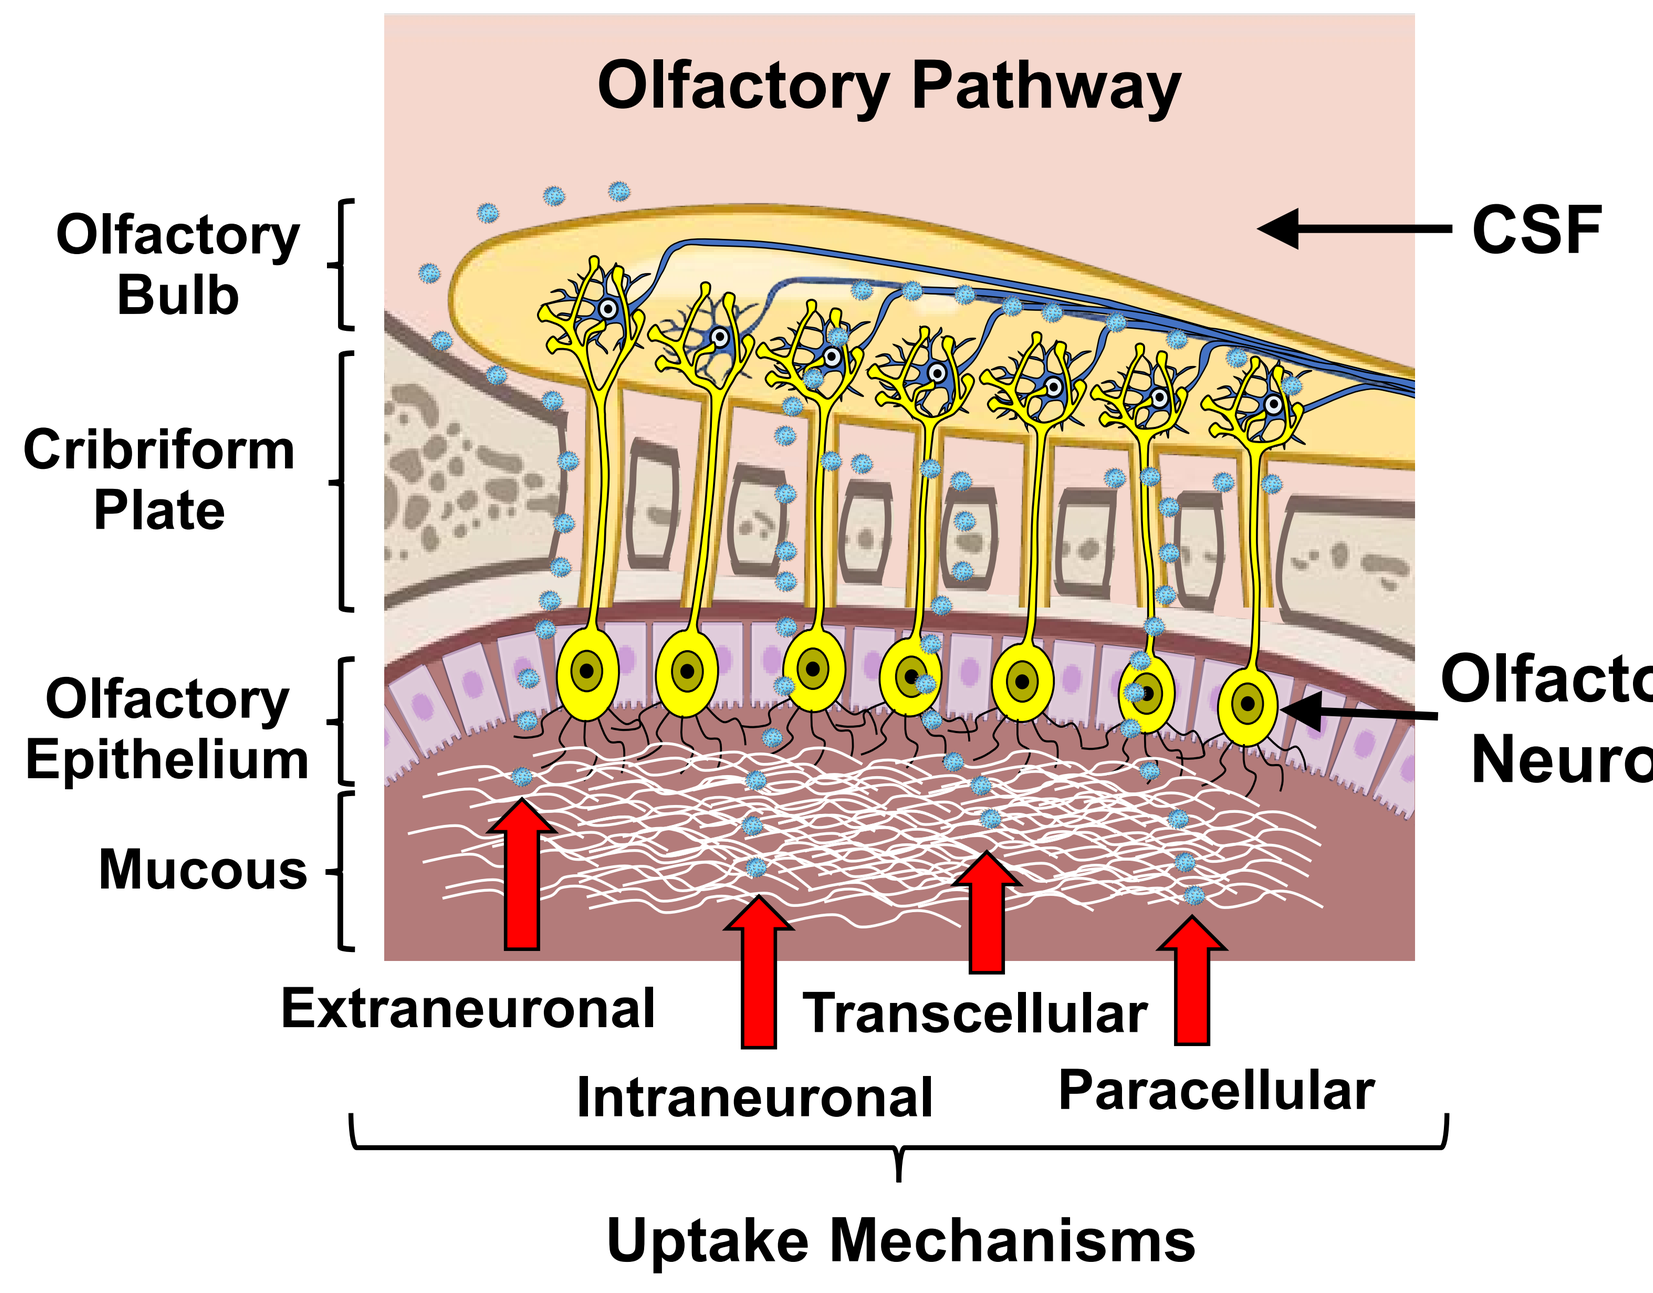
Olfactory Pathway
Olfactory Bulb
Cribriform Plate
Olfactory Epithelium
Mucous
CSF
Olfactory Neuron
Extraneuronal
Transcellular
Intraneuronal
Paracellular
Uptake Mechanisms

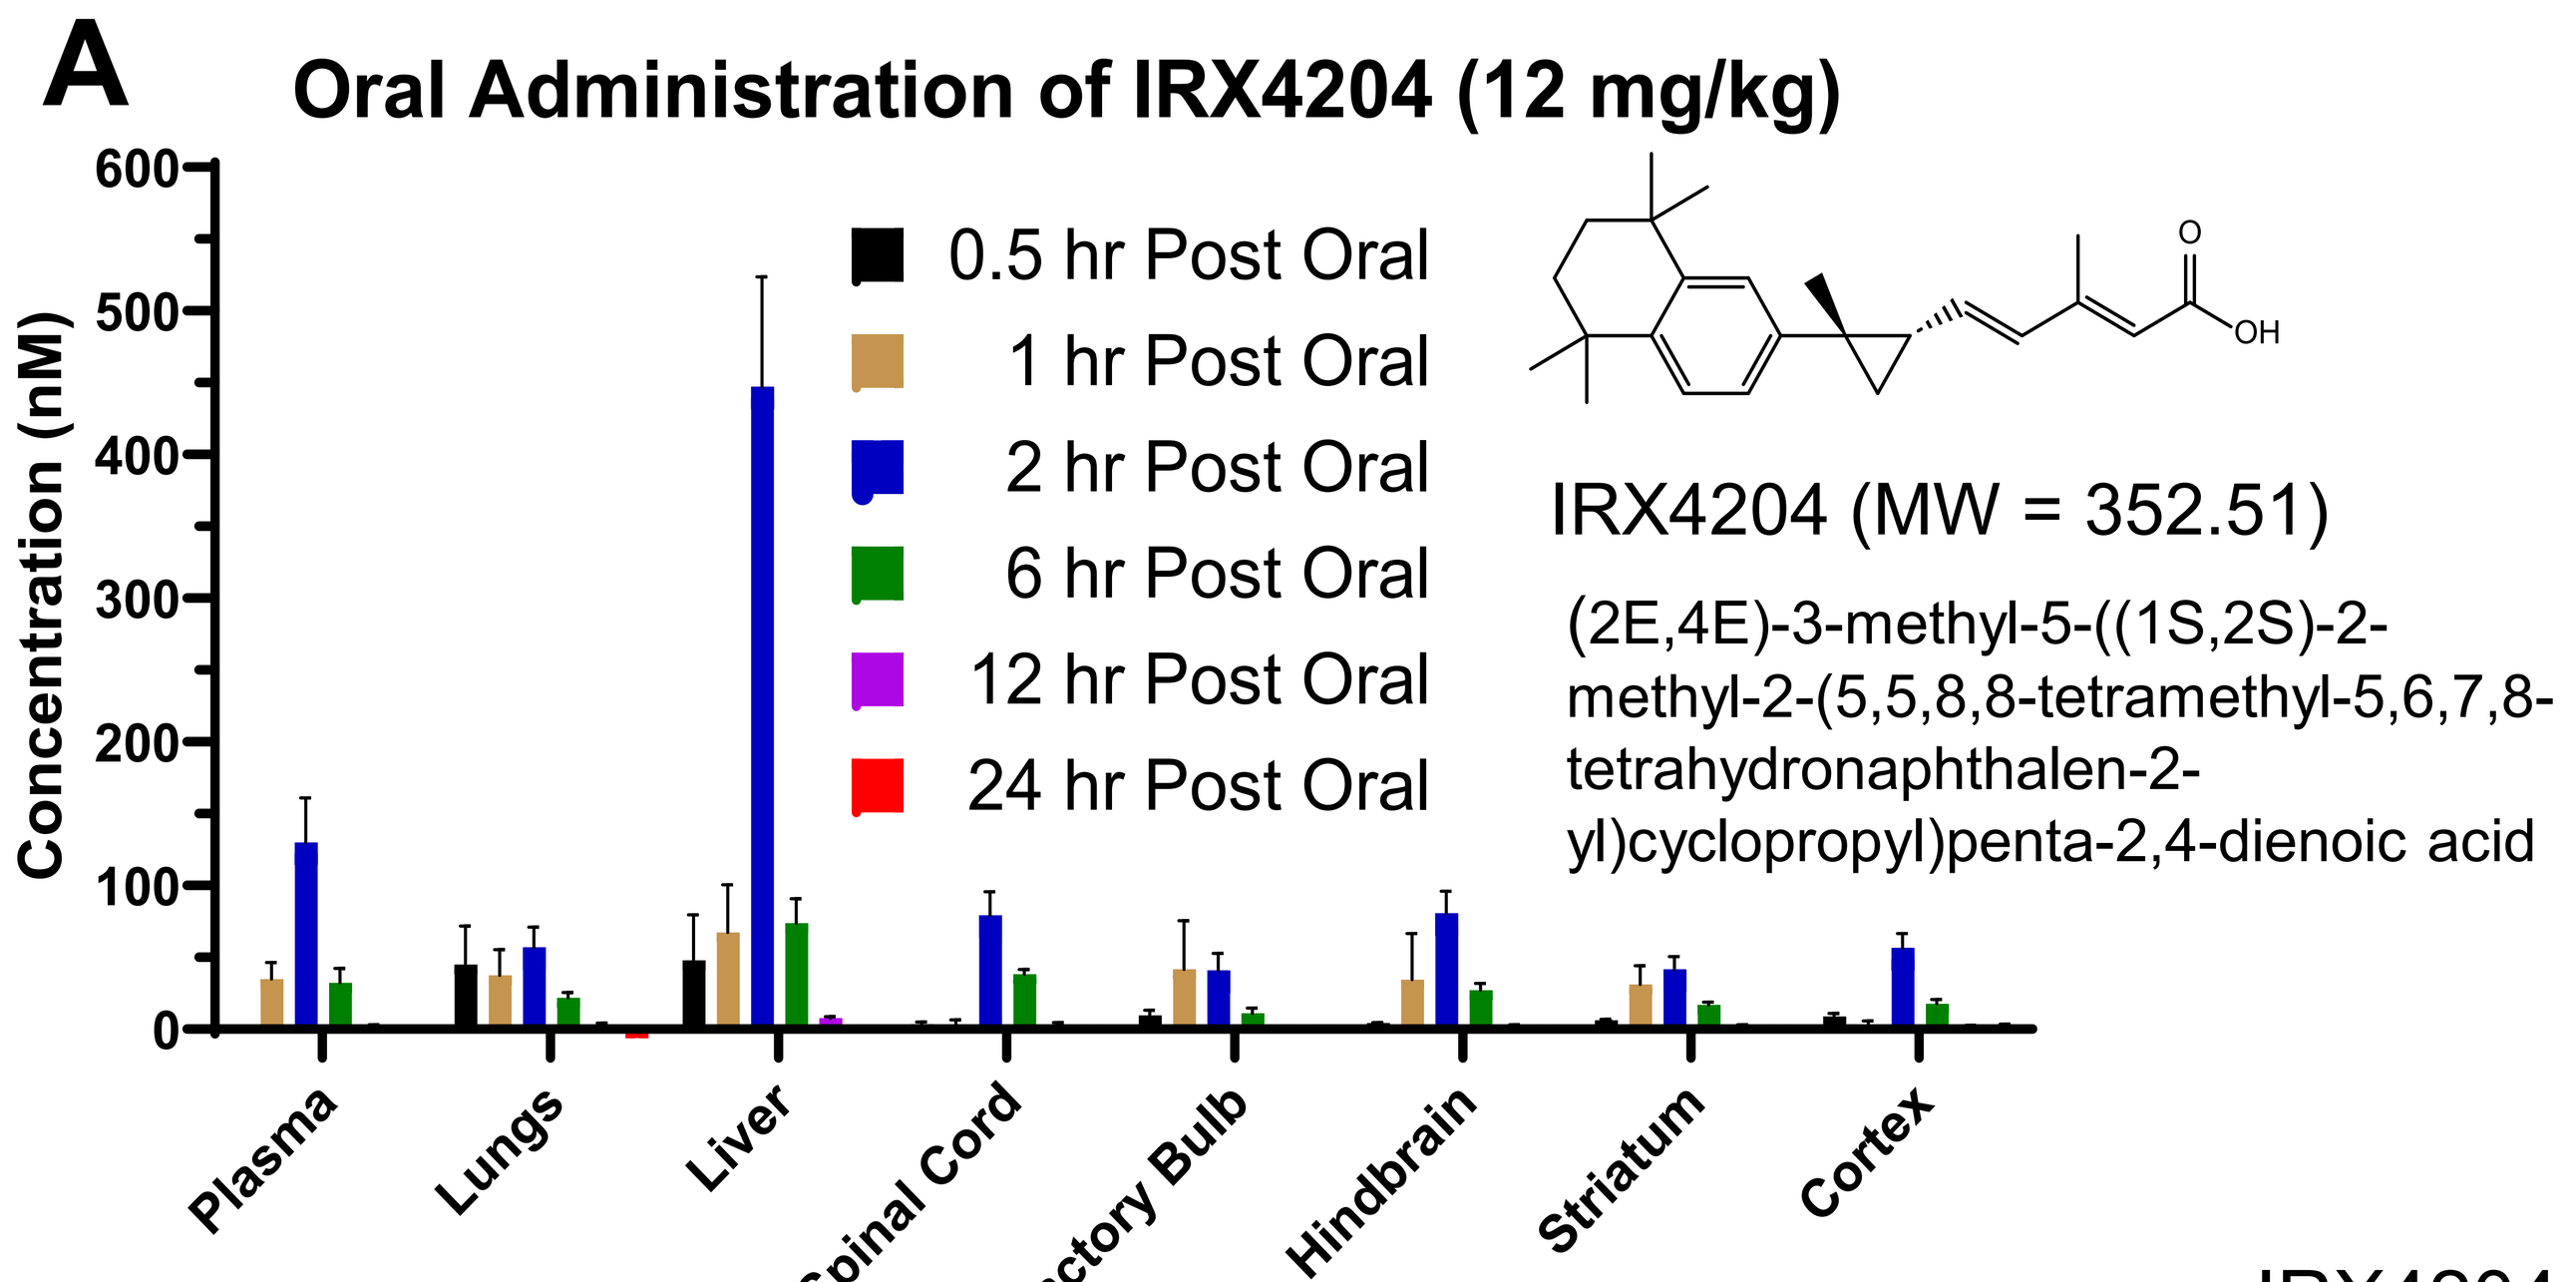
A
Oral Administration of IRX4204 (12 mg/kg)
Concentration (nM)
0.5 hr Post Oral
1 hr Post Oral
2 hr Post Oral
6 hr Post Oral
12 hr Post Oral
24 hr Post Oral
IRX4204 (MW = 352.51)
(2E,4E)-3-methyl-5-((1S,2S)-2-methyl-2-(5,5,8,8-tetramethyl-5,6,7,8-tetrahydronaphthalen-2-yl)cyclopropyl)penta-2,4-dienoic acid
Plasma
Lungs
Liver
Spinal Cord
Olfactory Bulb
Hindbrain
Striatum
Cortex

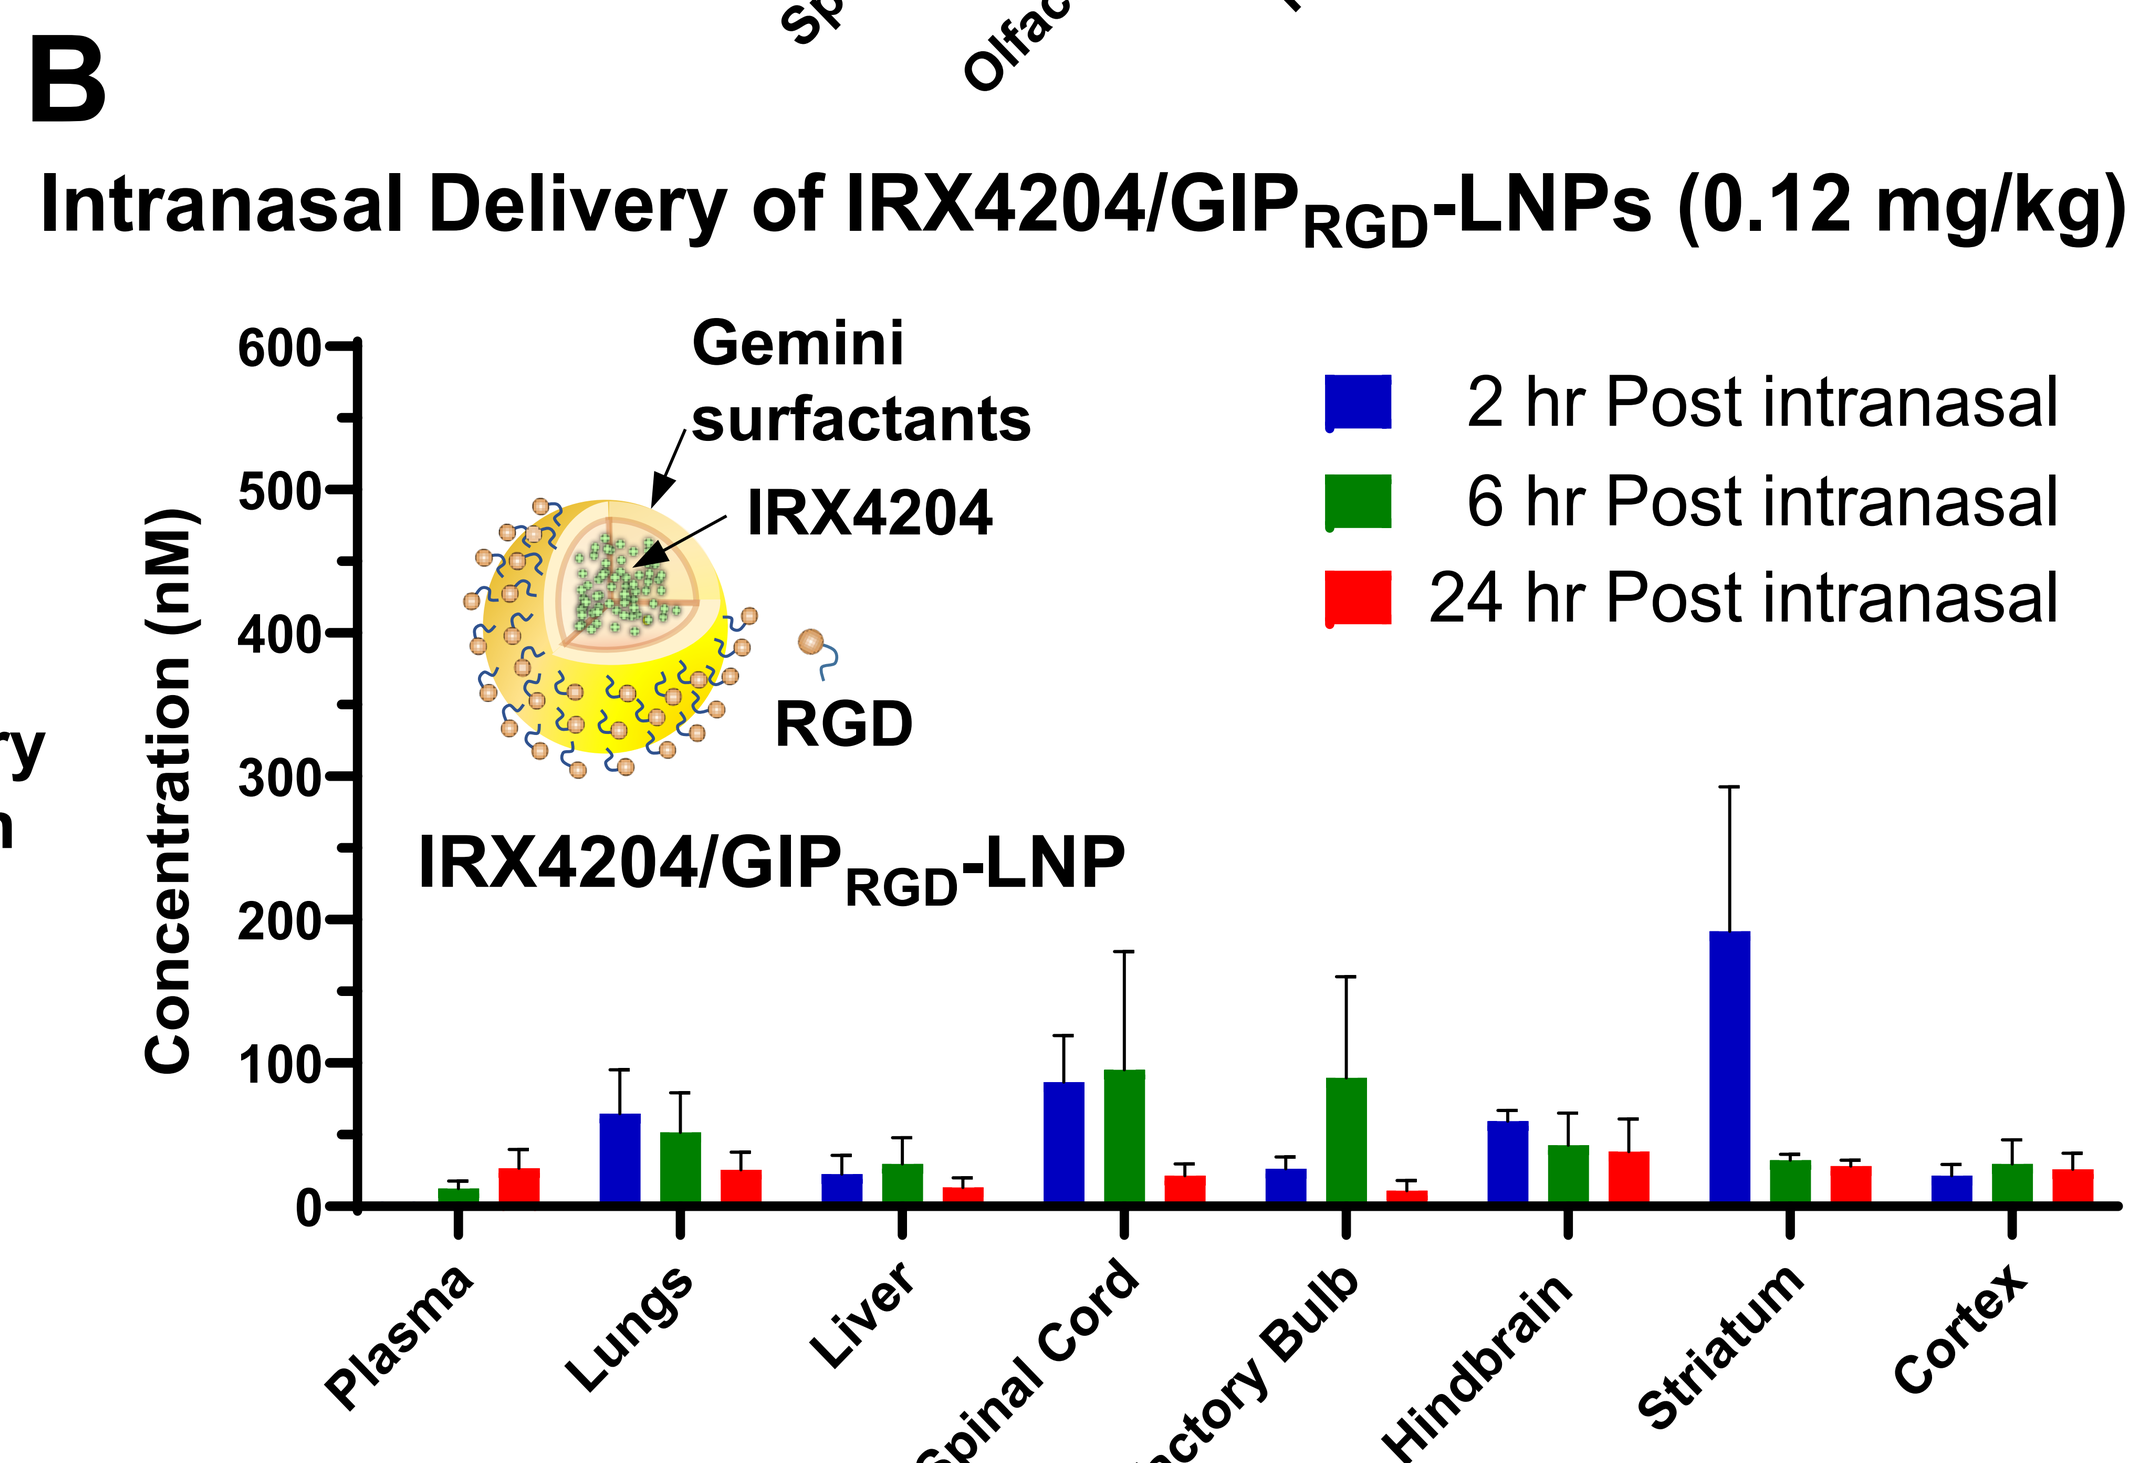
B
Intranasal Delivery of IRX4204/GIPRGD-LNPs (0.12 mg/kg)
Concentration (nM)
Gemini surfactants
IRX4204
RGD
IRX4204/GIPRGD-LNP
2 hr Post intranasal
6 hr Post intranasal
24 hr Post intranasal
Plasma
Lungs
Liver
Spinal Cord
Olfactory Bulb
Hindbrain
Striatum
Cortex

IRX4204 concentrations in the plasma, lungs, liver, spinal cord, olfactory bulb, hindbrain, striatum, and cortex of adult (20 g) female C57Bl/6 mice at varying times after a single oral gavage of IRX4204 (12 mg/kg) (A) or intranasal administration of IRX4204/GIP_{RGD}-LNPs corresponding to an IRX4204 dose of 0.12 mg/kg (B).

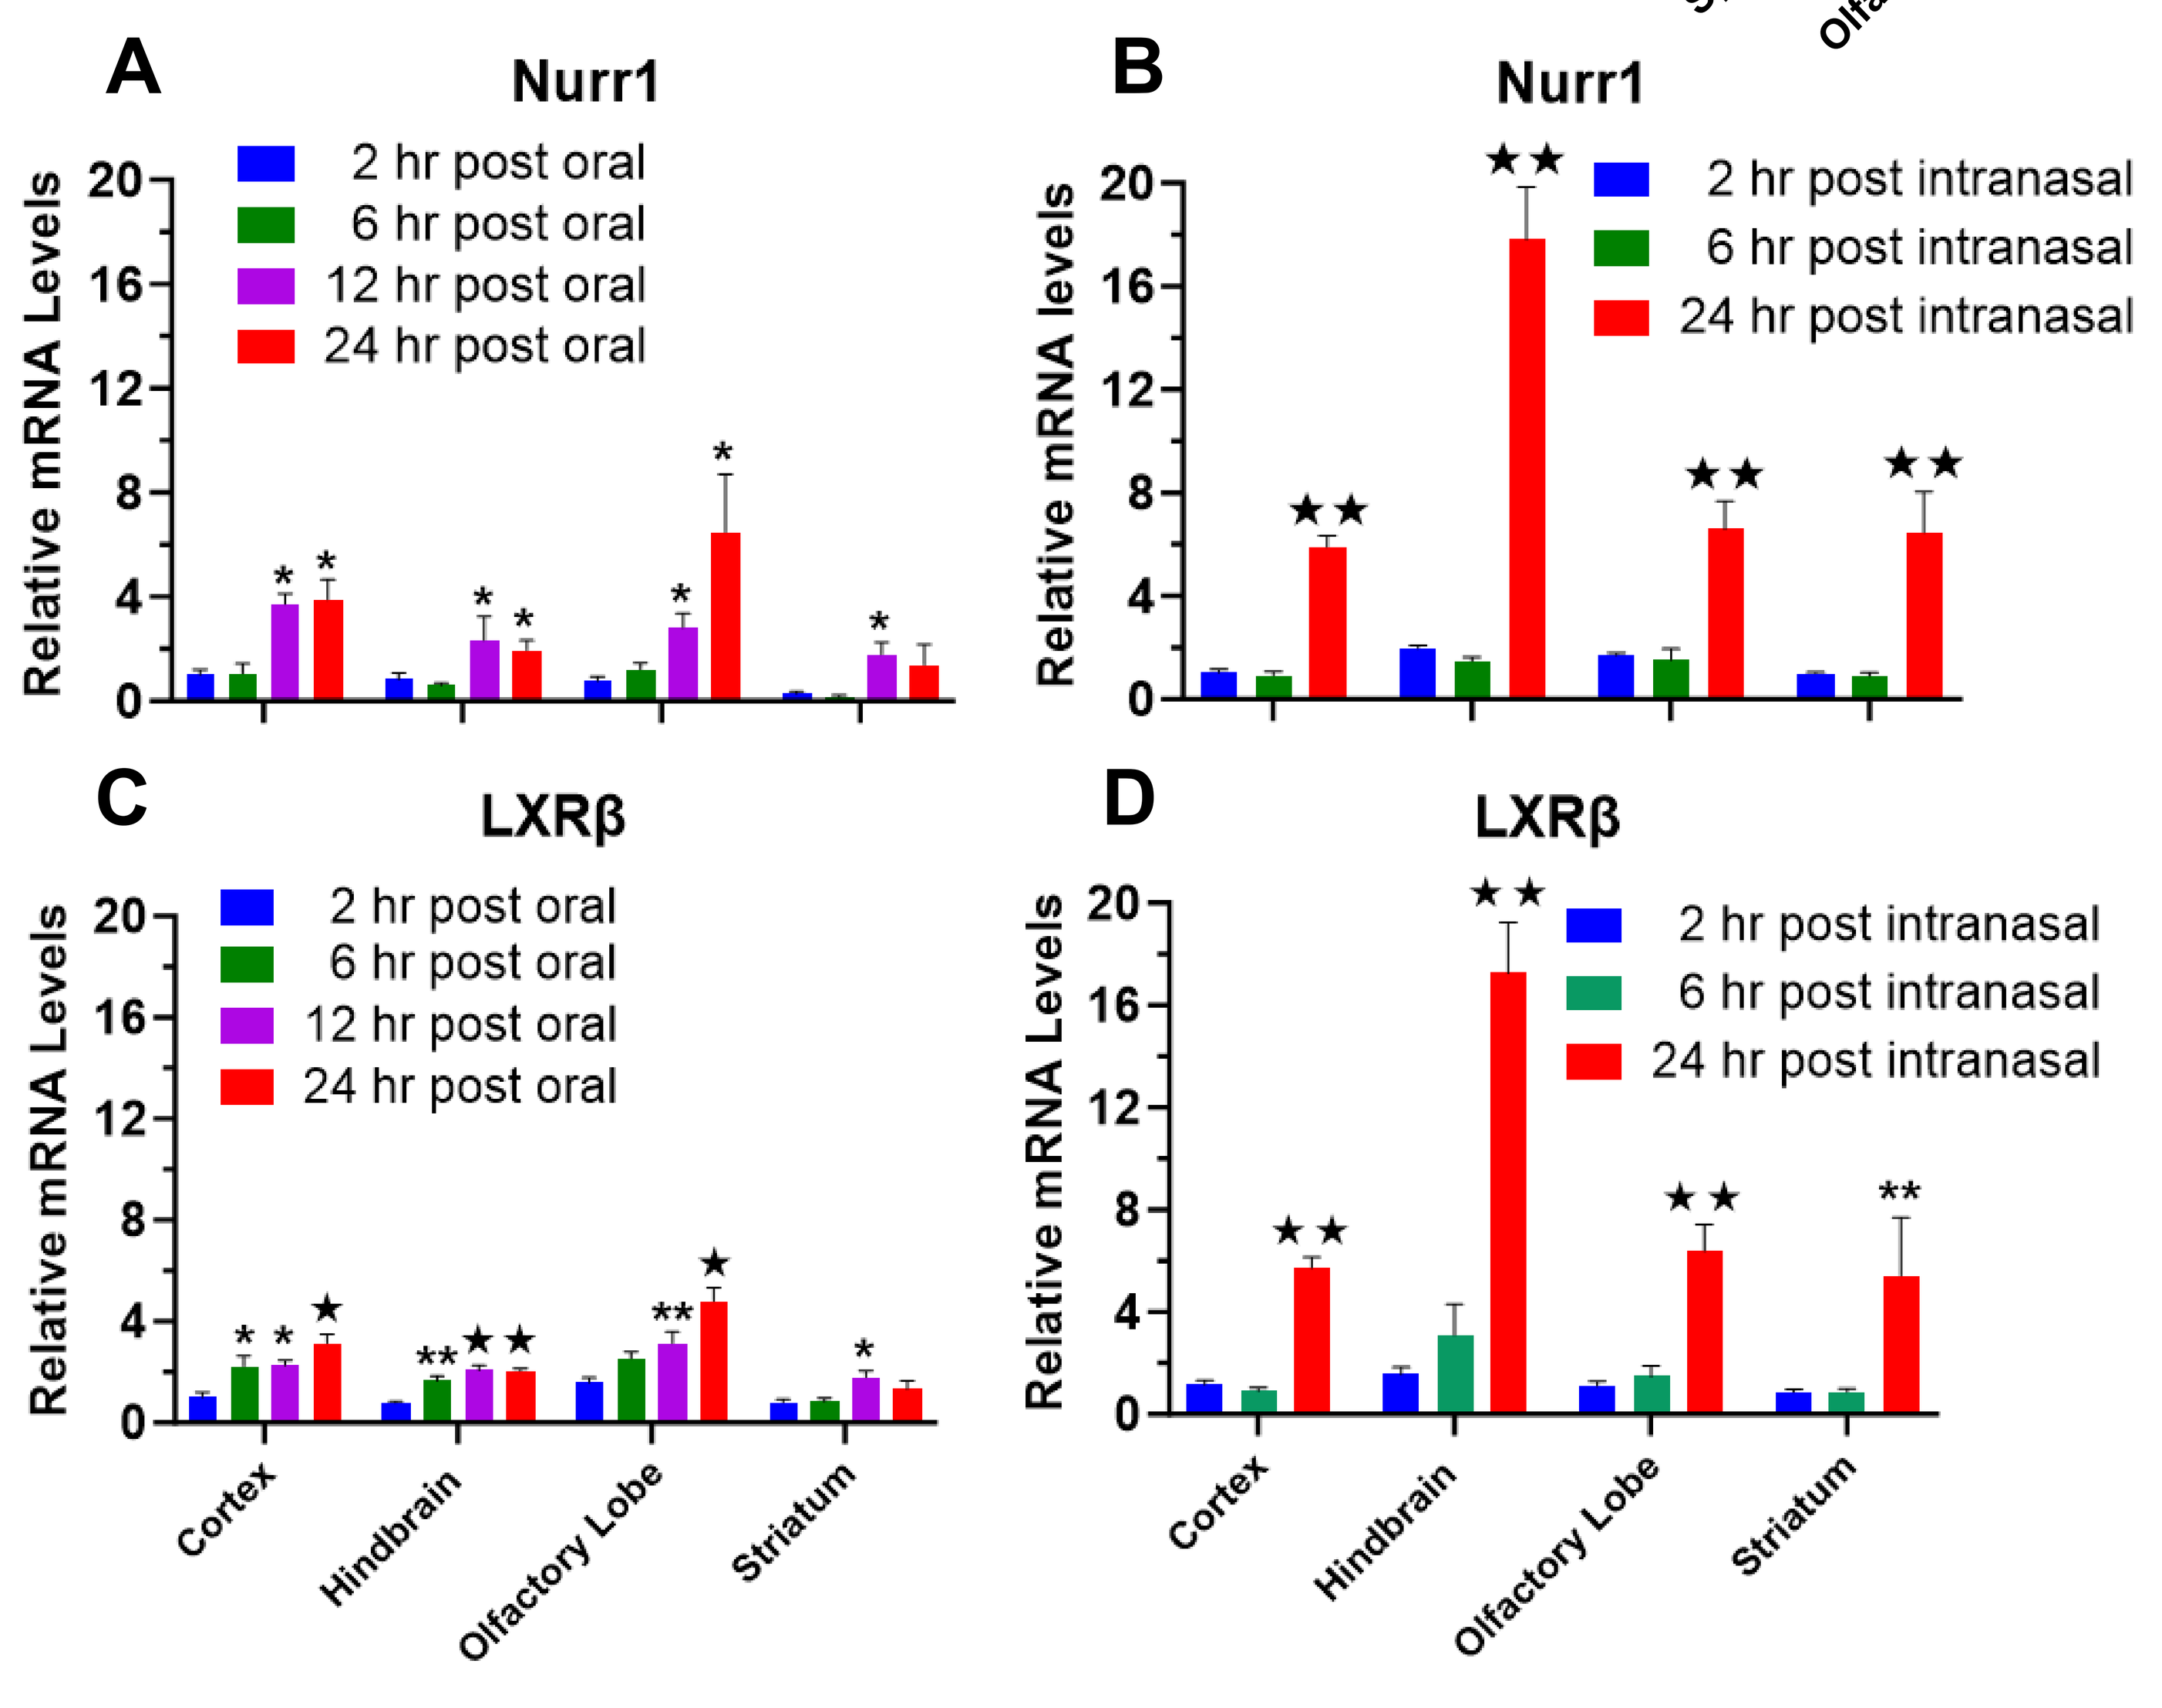
A
Nurr1
Relative mRNA Levels
2 hr post oral
6 hr post oral
12 hr post oral
24 hr post oral
B
Nurr1
Relative mRNA levels
2 hr post intranasal
6 hr post intranasal
24 hr post intranasal
C
LXRβ
Relative mRNA Levels
2 hr post oral
6 hr post oral
12 hr post oral
24 hr post oral
D
LXRβ
Relative mRNA Levels
2 hr post intranasal
6 hr post intranasal
24 hr post intranasal
Cortex
Hindbrain
Olfactory Lobe
Striatum

Nurr1 and LXRβ mRNA levels at varying times after the oral administration of IRX4204 (12 mg/kg; A and C) or intranasal delivery of IRX4204/GIP_{RGD}-LNPs (0.12 mg/kg; B and D) in female C57Bl/6 mice. Nurr1 and LXRβ mRNA levels are expressed relative to mice that were treated orally with vehicle (NEOBEE, 100 µl; A and C) or intranasally with empty GIP_{RGD}-LNPs suspended in water (24 µl; two installations of 6 µl into each nostril; B and D). *p<0.05, **p<0.01, ⋆p<0.001, and ⋆⋆p<0.0001 relative to the 2 hr time point.