Investigation of the RXR-Specific Agonist IRX4204 as a Disease-Modifying Agent of Alzheimer's Disease Neuropathology and Cognitive Impairment

MOUNT SINAI SCHOOL OF MEDICINE

Jun Wang[1,4], Martin Sanders[2], Rosh Chandraratna[2], Prashant Vempati[1], Weina Bi[1], Bing Gong[1], Wei Zhao[1], Kenjiro Ono[3], Masahito Yamada[3], Giulio Maria Pasinetti[1,4]*

[1]Department of Neurology, Mount Sinai School of Medicine, New York, New York 10029; [2]Io Therapeutics, Inc., 25241 Buckskin Drive, Laguna Hills, CA 92653. [3]Department of Neurology and Neurobiology of Aging, Kanazawa University Graduate School of Medical Science ,Kanazawa, Japan. [4]Geriatric Research, Education and Clinical Center, James J. Peters Veterans Affairs Medical Center, Bronx, New York 10468.

[] Address Correspondance to Giulio Maria Pasinetti: giulio.pasinetti@mssm.edu*

Background

Alzheimer's disease (AD) is characterized neuropathologically by the accumulation of extracellular plaques composed of Aβ protein, intracellular neurofibrillary tangles of hyperphosphorylated tau protein, and neuron loss. A major hypothesis for the pathogenesis of AD is that abnormally elevated Aβ contents in the brain of AD patients leads to the formation of insoluble Aβ fibrillar aggregates, which are the major constituents of senile plaques associated with neuronal loss in AD. The amyloid hypothesis is supported by substantial genetic and preclinical evidence including treatment effects of passive immunotherapies in mouse models of AD. Nonetheless, growing evidence suggests that cognitive deterioration in AD is directly linked to the accumulation of extracellular soluble oligomeric Aβ species rather than amyloid plaque deposition in the brain. Oligomeric Aβ, including high molecular weight (HMW) oligomeric Aβ as well as low-n oligomers from dimers to octamers, induces synapse degeneration, synaptic plasticity disruption and decreased long-term potentiation (LTP) , all of which contribute to mechanisms underlying the onset and progression of dementia in AD. As the accumulation of Aβ and various oligomeric forms of Aβ appears to be critical to AD dementia, therapies aiming to inhibit Aβ oligomerization are actively pursued. Recently, Bexarotene, a first-generation rexinoid, was shown to reduce Aβ neuropathology and cognitive impairments through mechanisms associated with ApoE-mediated clearance of Aβ in animal models of AD. IRX4204 is a second generation rexinoid with a higher potency and specificity to RXRs than Bexarotene. It is more selective for the RXRs and, as a consequence, it is associated with fewer adverse events in clinical applications.

Oral Administration of IRX4204 is Well Tolerated in TgCRND8 Mice

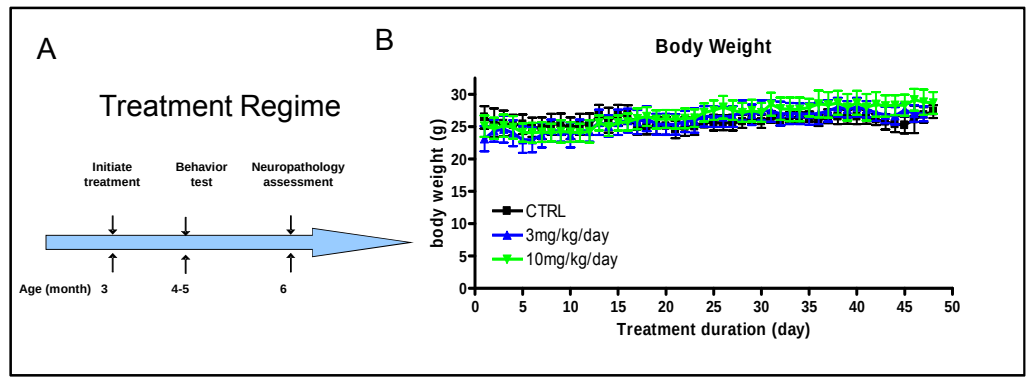

A
B
Treatment Regime
Initiate treatment
Behavior test
Neuropathology assessment
Age (month) 3
4-5
6
Body Weight
body weight (g)
CTRL
3mg/kg/day
10mg/kg/day
Treatment duration (day)

Short-term pilot study of the effects of IRX4204 *(A) Treatment regime: TgCRND8 mice were treated with 3 mg/kg/day or 10 mg/kg/day IRX4204 by daily gavage at 3 months of age, right at the time of the onset of the disease phenotype. Spatial memory tests were performed after 6 weeks of treatment followed by the contextual fear conditioning test. Brain neuropathology is analyzed at 6 months of age. (B) Body weight following gavage treatment*

IRX4204 Treatment Improves Cognitive Function

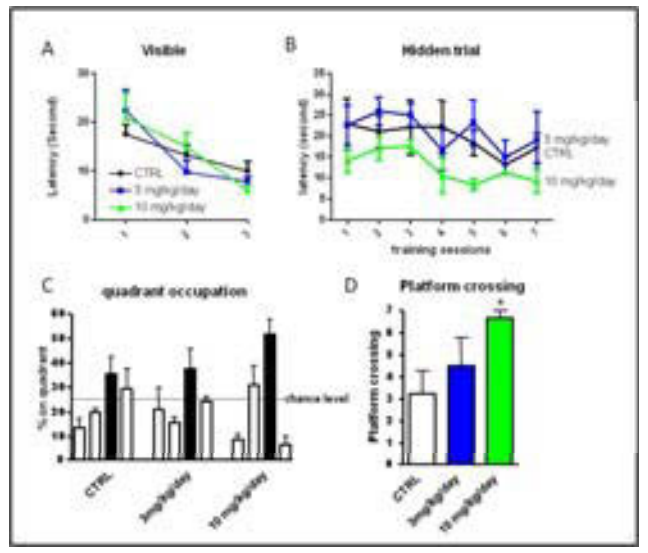

A
Visible
B
Hidden trial
C
quadrant occupation
D
Platform crossing

IRX4204 treatment improves spatial memory function by Morris water maze test in TgCRND8 mice *TgCRND8 mice were tested for cognitive function using the Morris water maze protocol (A) cued-visible trial (B) hidden learning trial: Acquisition of spatial memory is reflected by reducing escape latency time with increasing number of learning trials. (C, D) Probe trial: Spatial memory retention was assessed by measuring the % of time the animal spent in target quadrant (■ target quadrant) (C) or crossing the hidden platform (D).*

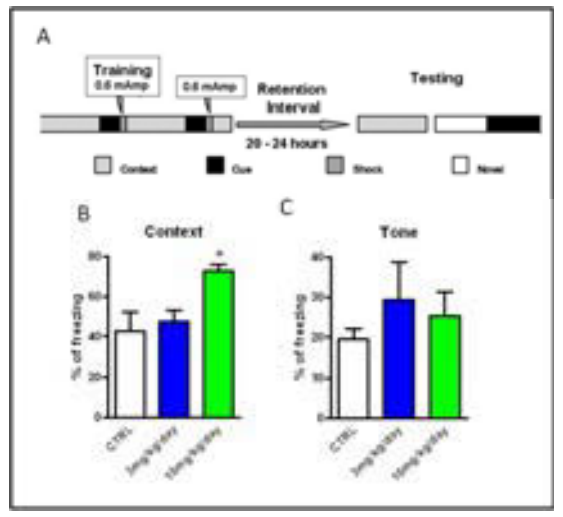

A
Training
Retention Interval
Testing
20 - 24 hours
B
Context
C
Tone

IRX4204 treatment improves cognition by contextual fear conditioning test in TgCRND8 mice *(A) contextual fear conditioning protocol: testing mouse is allowed to explore the chamber for 2 min. followed by an auditory cue for 15 s. The 2 s footshock is administered at the final 2 s of the CS. This procedure is repeated, and the testing mouse is returned to the home cage. Twenty hours after training, the testing mouse is returned to the same chamber in which training occurred (context), and freezing behavior is recorded. At the end of the 5 min context test, testing mouse is returned to the home cage. Approximately 1 h later, the testing mouse is introduced to a novel environment (newly modified the chamber) and freezing is recorded for 3 min. (B) Conditioned freezing during the Context test. (C) Conditioned freezing during the Tone test.*

IRX4204 Interferes with ß-Amyloid Peptide-to-Peptide Interactions

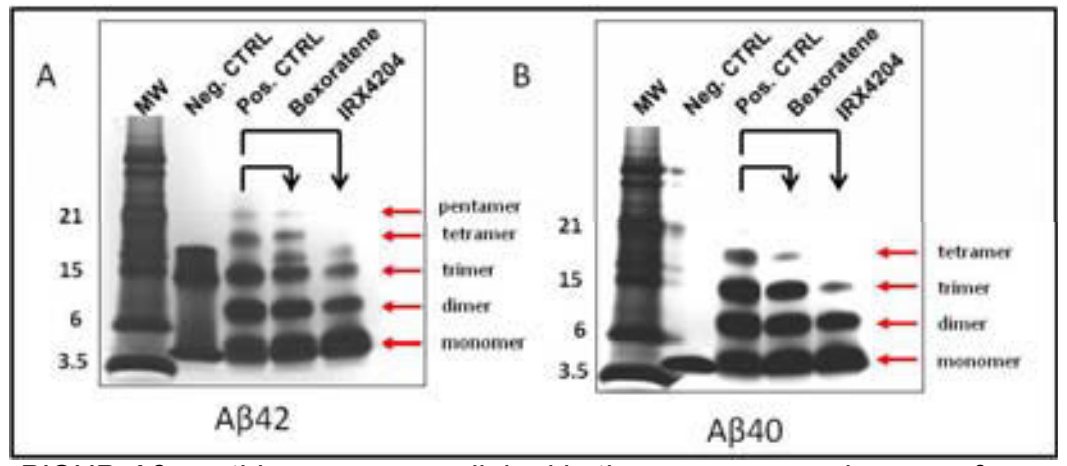

A
MW
Neg. CTRL
Pos. CTRL
Bexoratene
IRX4204
21
15
6
3.5
pentamer
tetramer
trimer
dimer
monomer
Aβ42
B
MW
Neg. CTRL
Pos. CTRL
Bexoratene
IRX4204
21
15
6
3.5
tetramer
trimer
dimer
monomer
Aβ40

IRX4204 interferes with Aβ oligomerization by Photo-induced cross-linking of unmodified proteins assay (PICUP) *(A) SDS-PAGE of Aβ$_{1-42}$ and (B) Aβ$_{1-40}$ in the presence or absence of IRX4204 or Bexarotene following PICUP. Aβ peptides were cross-linked in the presence or absence of compound and the bands in subsequent SDS gels were visualized using silver staining. In (A) and (B): MW: molecular weight; Neg CTRL: non-cross-linked Aβ$_{1-42}$, Aβ$_{1-40}$; Pos. CTRL: cross-linked aggregated Aβ$_{1-42}$, Aβ$_{1-40}$; Bexarotene and IRX4204: in the presence of 4x molar excess compared to Aβ. The Aβ$_{1-42}$ trimer band has been shown to be an SDS-induced artifact.*

IRX4204 Restores Long Term Potentiation in Hippocampal Slices with Synaptic Dysfunction

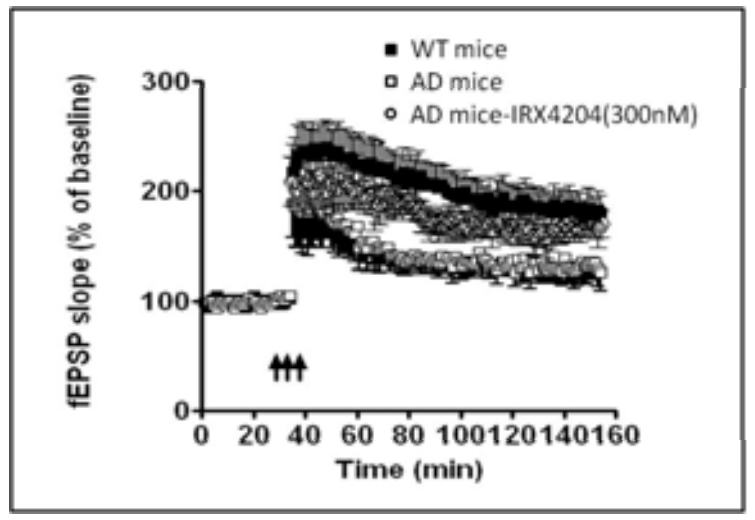

WT mice
AD mice
AD mice-IRX4204(300nM)
fEPSP slope (% of baseline)
Time (min)

The effect of IRX4204 on long term potentiation *Hippocampal slices (350 μm) isolated from old T2576 mice were acclimated in oxygenated artificial cerebrospinal fluid and treated with 300 nM IRX4204 for 1 hour. LTP was induced using theta-burst stimulation (four pulses at 100 Hz, with the bursts repeated at five Hz, and each tetanus including three 10-burst trains separated by 15 seconds) and fEPSPs were recorded from the CA1 region. The arrow indicates the beginning of tetanus to induce LTP, n=8 slices per group.*

IRX4204 Increases ApoE Expression in Primary Neurons

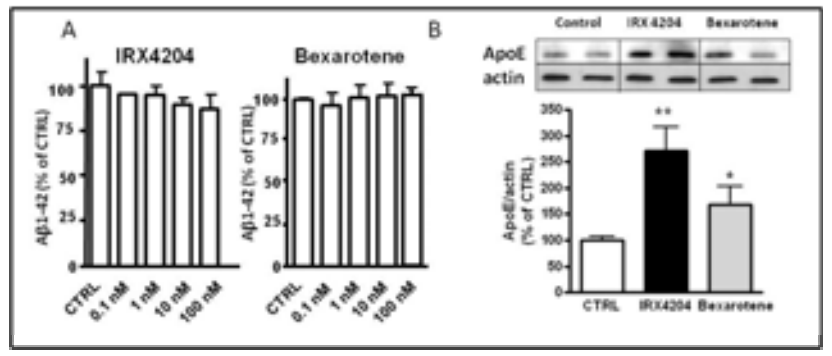

A
IRX4204
Bexarotene
Aβ1-42 (% of CTRL)
B
Control
IRX4204
Bexarotene
ApoE
actin
ApoE/actin (% of CTRL)
CTRL
IRX4204
Bexarotene

The effect of IRX4204 on ApoE expression in primary neurons *(A) The effect of IRX4204 on Aβ$_{1-42}$ generation in primary neurons derived from Tg2576 mice. Embryonic-day (E)16 cortico-hippocampal neuronal cultures were prepared from heterozygous Tg2576 transgenic mice (Tg2576 neurons) and seeded onto poly-D-lysine–coated 24-well plates at 5.0x 10^5 cells per well and cultured in Neurobasal medium supplemented with 2% B27, 0.5 mM L-glutamine and 1% penicillin-streptomycin. Neurons were treated with 0.1, 1, 10 or 100nM of IRX4204 (left panel) or Bexarotene (right panel) in quadruplets for 16 hours and conditioned medium was collected for Aβ detection.(B) The effect of IRX4204 on ApoE expression. Primary cortico-hippocampal neurons derived from C57B6 wild type mice were treated with 100nM of IRX4204 or Bexarotene or vehicle and total proteins were isolated and subjected to western blot analysis for ApoE expression (Data presented as mean±SEM, $^*p<0.05$, $^{**}P<0.01$, n=4 per group).*

Conclusions

•Our preliminary studies showed that short-term administration of IRX4204 can significantly improve cognitive function in the TgCRND8 mouse model of AD.
•The benefits of IRX4204 might be attributed to the anti-oligomerization activity of IRX4204, as well as its ability to improve LTP and possibly through mechanisms involving ApoE-mediated Aβ clearance